Discovery and Optimization of GPBAR1 (TGR5) Agonists

James R. Beasley, Joyce Huang, Christina Gallo-Ebert, Adam Bata, Melissa Donigan, Caroline Giordano, Nalini Kanneganti, John Malone, Ellen Sieber-McMaster, Stan Nawoschik, Linda O'Brien, Quynhchi Pham, Elena Severinova, Dongchuan Shi, Robert Swanson, Kiersten Tovar, Xiaoqing Xu, Petronio Zalamea, Joseph Nickels, Philip Stein, & Maria Webb

Abstract

Multiple series of GPBAR1 (TGR5) agonists were identified from an uHTS campaign using the ECLiPS Technology. One series has emerged during the optimization to possess potent full agonist activity in cAMP assays, initiate GLP-1 secretion in H716 cells, and show improved glucose tolerance in *in vivo* mouse studies. This novel GPBAR1 agonist series are potentially useful therapeutics for type II diabetes.

GPBAR1 is the Bile Acid GPCR

Type II diabetes is recognized as a major health threat in the world today. Sufferers of type II diabetes do not have sufficient insulin levels or sensitivity resulting in hyperglycemia that can have life-threatening consequences if left untreated. One way to increase insulin levels and/or sensitivity is to increase the level of glucagon like peptide-1 (GLP-1).

GPBAR1 (TGR5) is a G_s-coupled bile acid receptor shown to regulate a number of metabolic processes, including secretion of GLP-1 from intestinal L-cells. Activation of GPBAR1 in intestinal L-cells with an agonist initiates the cAMP cascade resulting in the secretion of GLP-1.

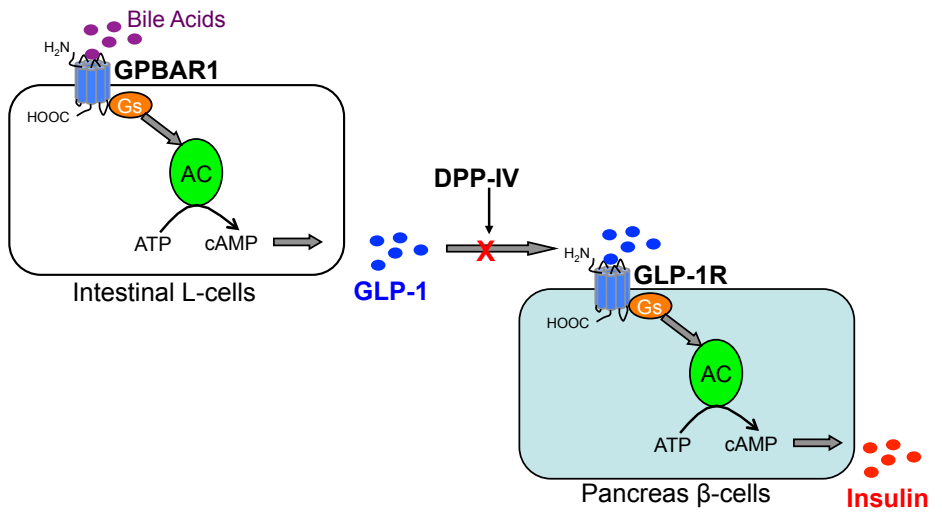
Bile Acids
H_2N
GPBAR1
HOOC
Gs
AC
ATP
cAMP
Intestinal L-cells
GLP-1
DPP-IV
H_2N
GLP-1R
HOOC
Gs
AC
ATP
cAMP
Pancreas β-cells
Insulin

Multiple Series Found in uHTS

GPBAR1 was screened in 1536-well format using Venenum Biodesign's ECLiPS technology. The survey of the nearly 6 million member compound collection identified multiple GPBAR1-specific non-bile like agonistic chemotypes with potencies $\leq$ 5 μM.

Optimization has led to Improved Properties

Multiple series of GPBAR1 agonists were examined for their properties with one series emerging as a clear front-runner. This series of non-bile acid-like small molecules was discovered as a 328 nM hit from the ECLiPS Screening Collection. Optimization of the structure led to low nM potency.

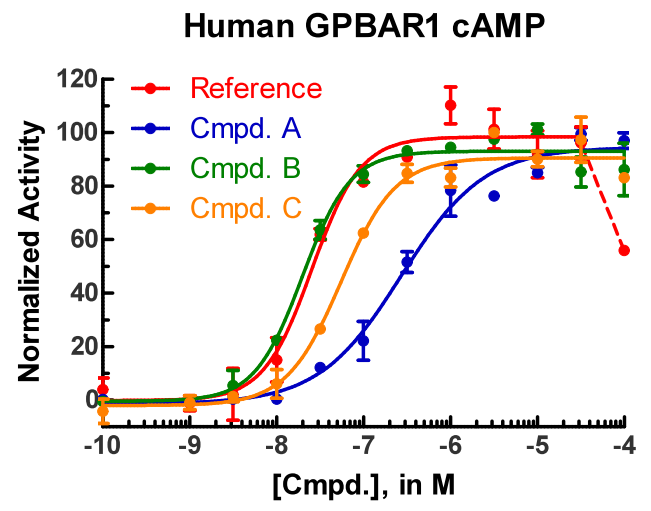
Human GPBAR1 cAMP
Reference
Cmpd. A
Cmpd. B
Cmpd. C
Normalized Activity
120
100
80
60
40
20
0
-10
-9
-8
-7
-6
-5
-4
[Cmpd.], in M

One of the largest issues with previous GPBAR1 agonists is the absence of activity against the rodent GPBAR1. Our lead series shows potent activity against the mouse receptor thus allowing for *in vivo* modeling of the compounds to be done in mice instead of other more expensive mammalian models (Table below).

Another important property of this series is the lack of activity towards the soluble bile-acid receptor FXR (Table below). Avoiding FXR activity could be crucial in avoiding potential side effects.

Cmpd	Ref	A	B	C
hGPBAR1 (CHO, cAMP)	26	328	24	47
mGPBAR1 (HEK, cAMP)	4	3,140	94	96
NCI-H716, (cAMP)	60	1,760	71	128
FXR Co-activation	>100,000	-	>100,000	>100,000
Solubility	<1,000	2,000	5,000	85,000

All numbers in nM

Agonist causes GLP-1 Secretion

NCI-H716 is a human intestinal cell line with expression of GPBAR1 which has the ability to secrete GLP-1. GPBAR1 agonists that cause a cAMP response in NCI-H716 were further tested to determine their ability to cause GLP-1 secretion. Both compound B and the literature reference demonstrated the ability to cause GLP-1 secretion.

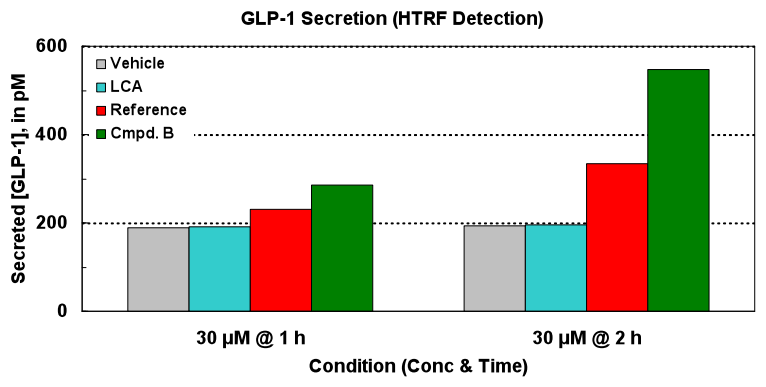
GLP-1 Secretion (HTRF Detection)
Vehicle
LCA
Reference
Cmpd. B
Secreted [GLP-1], in pM
600
400
200
0
30 μM @ 1 h
30 μM @ 2 h
Condition (Conc & Time)

Agonist shows *In Vivo* Activity

Diet-induced obese (DIO) mice were dosed with compound via intracolonic injection of the small molecule agonists at 30 mg/kg. After recovery, an oral glucose tolerance test was administered. Both compound B and the literature reference demonstrated enhanced glucose clearance consistent with elevated GLP-1 levels.

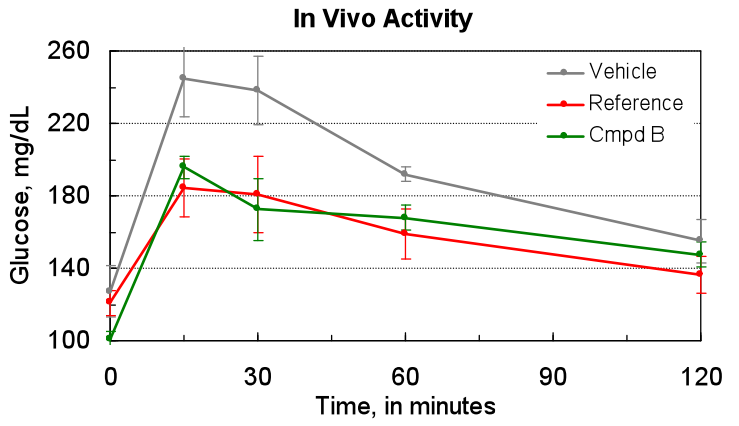
In Vivo Activity
Vehicle
Reference
Cmpd B
Glucose, mg/dL
260
220
180
140
100
0
30
60
90
120
Time, in minutes

Conclusion

A novel series of GPBAR1 agonists has been discovered. This series of potent and selective compounds has shown the ability to induce GLP-1 secretion in H716 cells and is efficacious in an acute mouse model via intracolonic injection. This shows that these GPBAR1 agonists are potentially useful therapeutic agents for metabolic disorders such as Type II diabetes.